P-969

Differences in Demographics and HIV Risk Exposures Among People Receiving Bictegravir/Emtricitabine/Tenofovir Alafenamide (B/F/TAF) Versus Other Post-Exposure Prophylaxis Regimens in the United States, 2016-2025

Kenneth H Mayer[1], Travis Lim[2], Valentina Shvachko[2], Laura M Mann[3], Wenyi Wang[2], Jason T Hindman[2]

[1]Fenway Health, Boston, MA, USA; [2]Gilead Sciences, Inc., Foster City, CA, USA; [3]IQVIA, Atlanta, GA, USA

Kenneth H Mayer
kmayer@fenwayhealth.org
Fenway Health
Boston, MA 02215, USA
Tel: +1 (617) 927-6087

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors

Conclusions

- A total of 94,018 post-exposure prophylaxis episodes were assessed, including prescriptions for raltegravir + emtricitabine/tenofovir disoproxil fumarate, dolutegravir + emtricitabine/tenofovir disoproxil fumarate, and bictegravir/emtricitabine/tenofovir alafenamide
 - — In addition, 117,053 post-exposure prophylaxis–eligible encounters with no associated post-exposure prophylaxis claim were identified
- Among episodes with linked exposure information, bictegravir/emtricitabine/tenofovir alafenamide was proportionally more often associated with rectal sexually transmitted infection testing codes and other proxy indicators of sexual HIV exposure risk
 - — This may reflect differences in prescribing settings, patient populations, coding practices, and/or uptake patterns during the study period
- Post-exposure prophylaxis prescriptions for bictegravir/emtricitabine/tenofovir alafenamide increased steadily from 2018 to 2024, even before guidelines formally changed to recommend it in 2025, although differences across regimens may have been influenced by calendar-time effects
 - — Post-exposure prophylaxis prescriptions for dolutegravir + emtricitabine/tenofovir disoproxil fumarate increased from 2016 to 2024, whereas prescriptions for raltegravir + emtricitabine/tenofovir disoproxil fumarate declined and then plateaued after 2019
- Among those with documented HIV-negative baseline testing, HIV diagnoses following post-exposure prophylaxis were rare across all regimens
- These data suggest that many HIV exposure–coded encounters had no observable associated post-exposure prophylaxis pharmacy claims and that HIV diagnoses following post-exposure prophylaxis were uncommon among individuals with documented baseline HIV-negative testing and observable follow-up
 - — These findings may help inform efforts to improve targeted post-exposure prophylaxis access and identify populations in whom HIV prevention services could be strengthened

Plain Language Summary

- Post-exposure prophylaxis uses medication to help prevent HIV infection after a person has been exposed to the virus
- People who could benefit from post-exposure prophylaxis do not always receive a prescription or complete the full course of treatment
- This study used health insurance records to look at patterns of post-exposure prophylaxis use and how many people were diagnosed with HIV after receiving post-exposure prophylaxis
- From 2016 to 2024, the use of bictegravir/emtricitabine/tenofovir alafenamide and dolutegravir + emtricitabine/tenofovir disoproxil fumarate increased, while the use of raltegravir + emtricitabine/tenofovir disoproxil fumarate decreased and then remained steady after 2019
- The use of bictegravir/emtricitabine/tenofovir alafenamide increased steadily after it became available. It was commonly used among people whose records included indicators consistent with sexual HIV exposure
- Very few people were diagnosed with HIV after receiving post-exposure prophylaxis
- These findings may help to identify populations and settings where HIV prevention services could be strengthened

Introduction

- Effective treatment and prevention strategies are required to address the ongoing HIV public health crisis in the United States[1]
- Post-exposure prophylaxis (PEP) entails a 28-day course of antiretroviral therapy (ART) started within <72 hours after a suspected exposure in an individual without evidence of HIV infection[2]
- Previously recommended PEP regimens, including raltegravir + emtricitabine/tenofovir disoproxil fumarate (RAL + F/TDF) and ritonavir-boosted darunavir + emtricitabine/tenofovir disoproxil fumarate (DRV/r + F/TDF), presented tolerability issues, twice-daily dosing requirements, and/or multi-tablet formulations leading to adherence challenges[3]
 - — In 2025, the Centers for Disease Control and Prevention (CDC) introduced updated recommendations for PEP, with the preferred regimens being dolutegravir + (tenofovir alafenamide OR tenofovir disoproxil fumarate) + (emtricitabine OR lamivudine) as a 2-pill, once-daily option and bictegravir/emtricitabine/tenofovir alafenamide (B/F/TAF) as a single-tablet, once-daily option[2]
- PEP uptake and completion rates remain suboptimal, with notable variation across populations, which may reflect gaps in awareness, acceptability, stigma, and access[4-6]
- The initial PEP encounter represents an opportunity for health care providers to initiate linkage to pre-exposure prophylaxis (PrEP) for patients continuously engaging in high-risk behaviors[7]
- Understanding the exposure situations and population characteristics driving PEP use is important for evaluating whether PEP is being prescribed as a targeted prevention strategy in higher-risk exposure circumstances

Objective

- To describe demographic characteristics, associated HIV exposure codes, prescribing patterns, and post-PEP HIV outcomes, with the goal of identifying opportunities to support and optimize PEP provision and access across populations, regions, and programs

Methods

- A retrospective observational study was conducted from April 2016 through September 2025, using US administrative claims data from HealthVerity Marketplace from as early as January 2015 to allow for a pre-index lookback period to capture historical claims
 - — The database covers >200 million individuals across private insurance, Medicaid, and Medicare with linked pharmacy, medical, and laboratory testing claims
- The study population included people aged ≥12 years who had medical records with nonmissing sex and year of birth and had an index date (date of first eligible PEP claim) on or after April 18, 2016
- HIV risk exposures were categorized using *International Classification of Diseases, Tenth Revision, Clinical Modification* diagnosis codes and laboratory testing codes linked to PEP episodes within <72 hours, including HIV exposure, occupational exposure, sexual exposure, and other exposures

For additional exposure and diagnosis code details, please scan the QR code

- PEP regimens were defined as 28- or 30-day supply claims for RAL + F/TDF, dolutegravir + emtricitabine/tenofovir disoproxil fumarate (DTG + F/TDF), B/F/TAF, and DRV/r + F/TDF and were identified based on a previously published approach using the algorithm shown in **Figure 1**[8]
- HIV diagnosis following PEP use was assessed during Days 28 to 42 post-PEP and was defined as an HIV diagnosis or reactive HIV test after PEP, with documented evidence that the individual was negative for HIV at baseline (–3 to +10 days relative to the index date)
- PEP trends by year were assessed for each regimen, regardless of whether an associated risk exposure was found within <72 hours
- Data were summarized using descriptive statistics, including counts, mean (SD), and median (IQR); all data were at the PEP episode level
- Statistical comparisons were performed using the chi-squared test for categorical variables; standardized mean differences were also calculated to assess the magnitude of differences between regimen groups

Results

Study Population and Demographic Characteristics

- The PEP algorithm (**Figure 1**) identified 83,623 individuals with 94,018 PEP episodes, with a median (IQR) of 1 (1-1) episode per person
 - — Among all PEP users, 7080 individuals had >1 PEP episode
 - — Among the 26,359 exposure-linked episodes, 11,981 were associated with RAL + F/TDF, 12,151 with DTG + F/TDF, and 2227 with B/F/TAF
- In contrast, 117,053 encounters were identified with ≥1 claim with exposure to HIV as the primary diagnosis code that were eligible for PEP (with no history of HIV or ART for treatment) but for which no associated PEP claims were found

Figure 1. PEP Algorithm

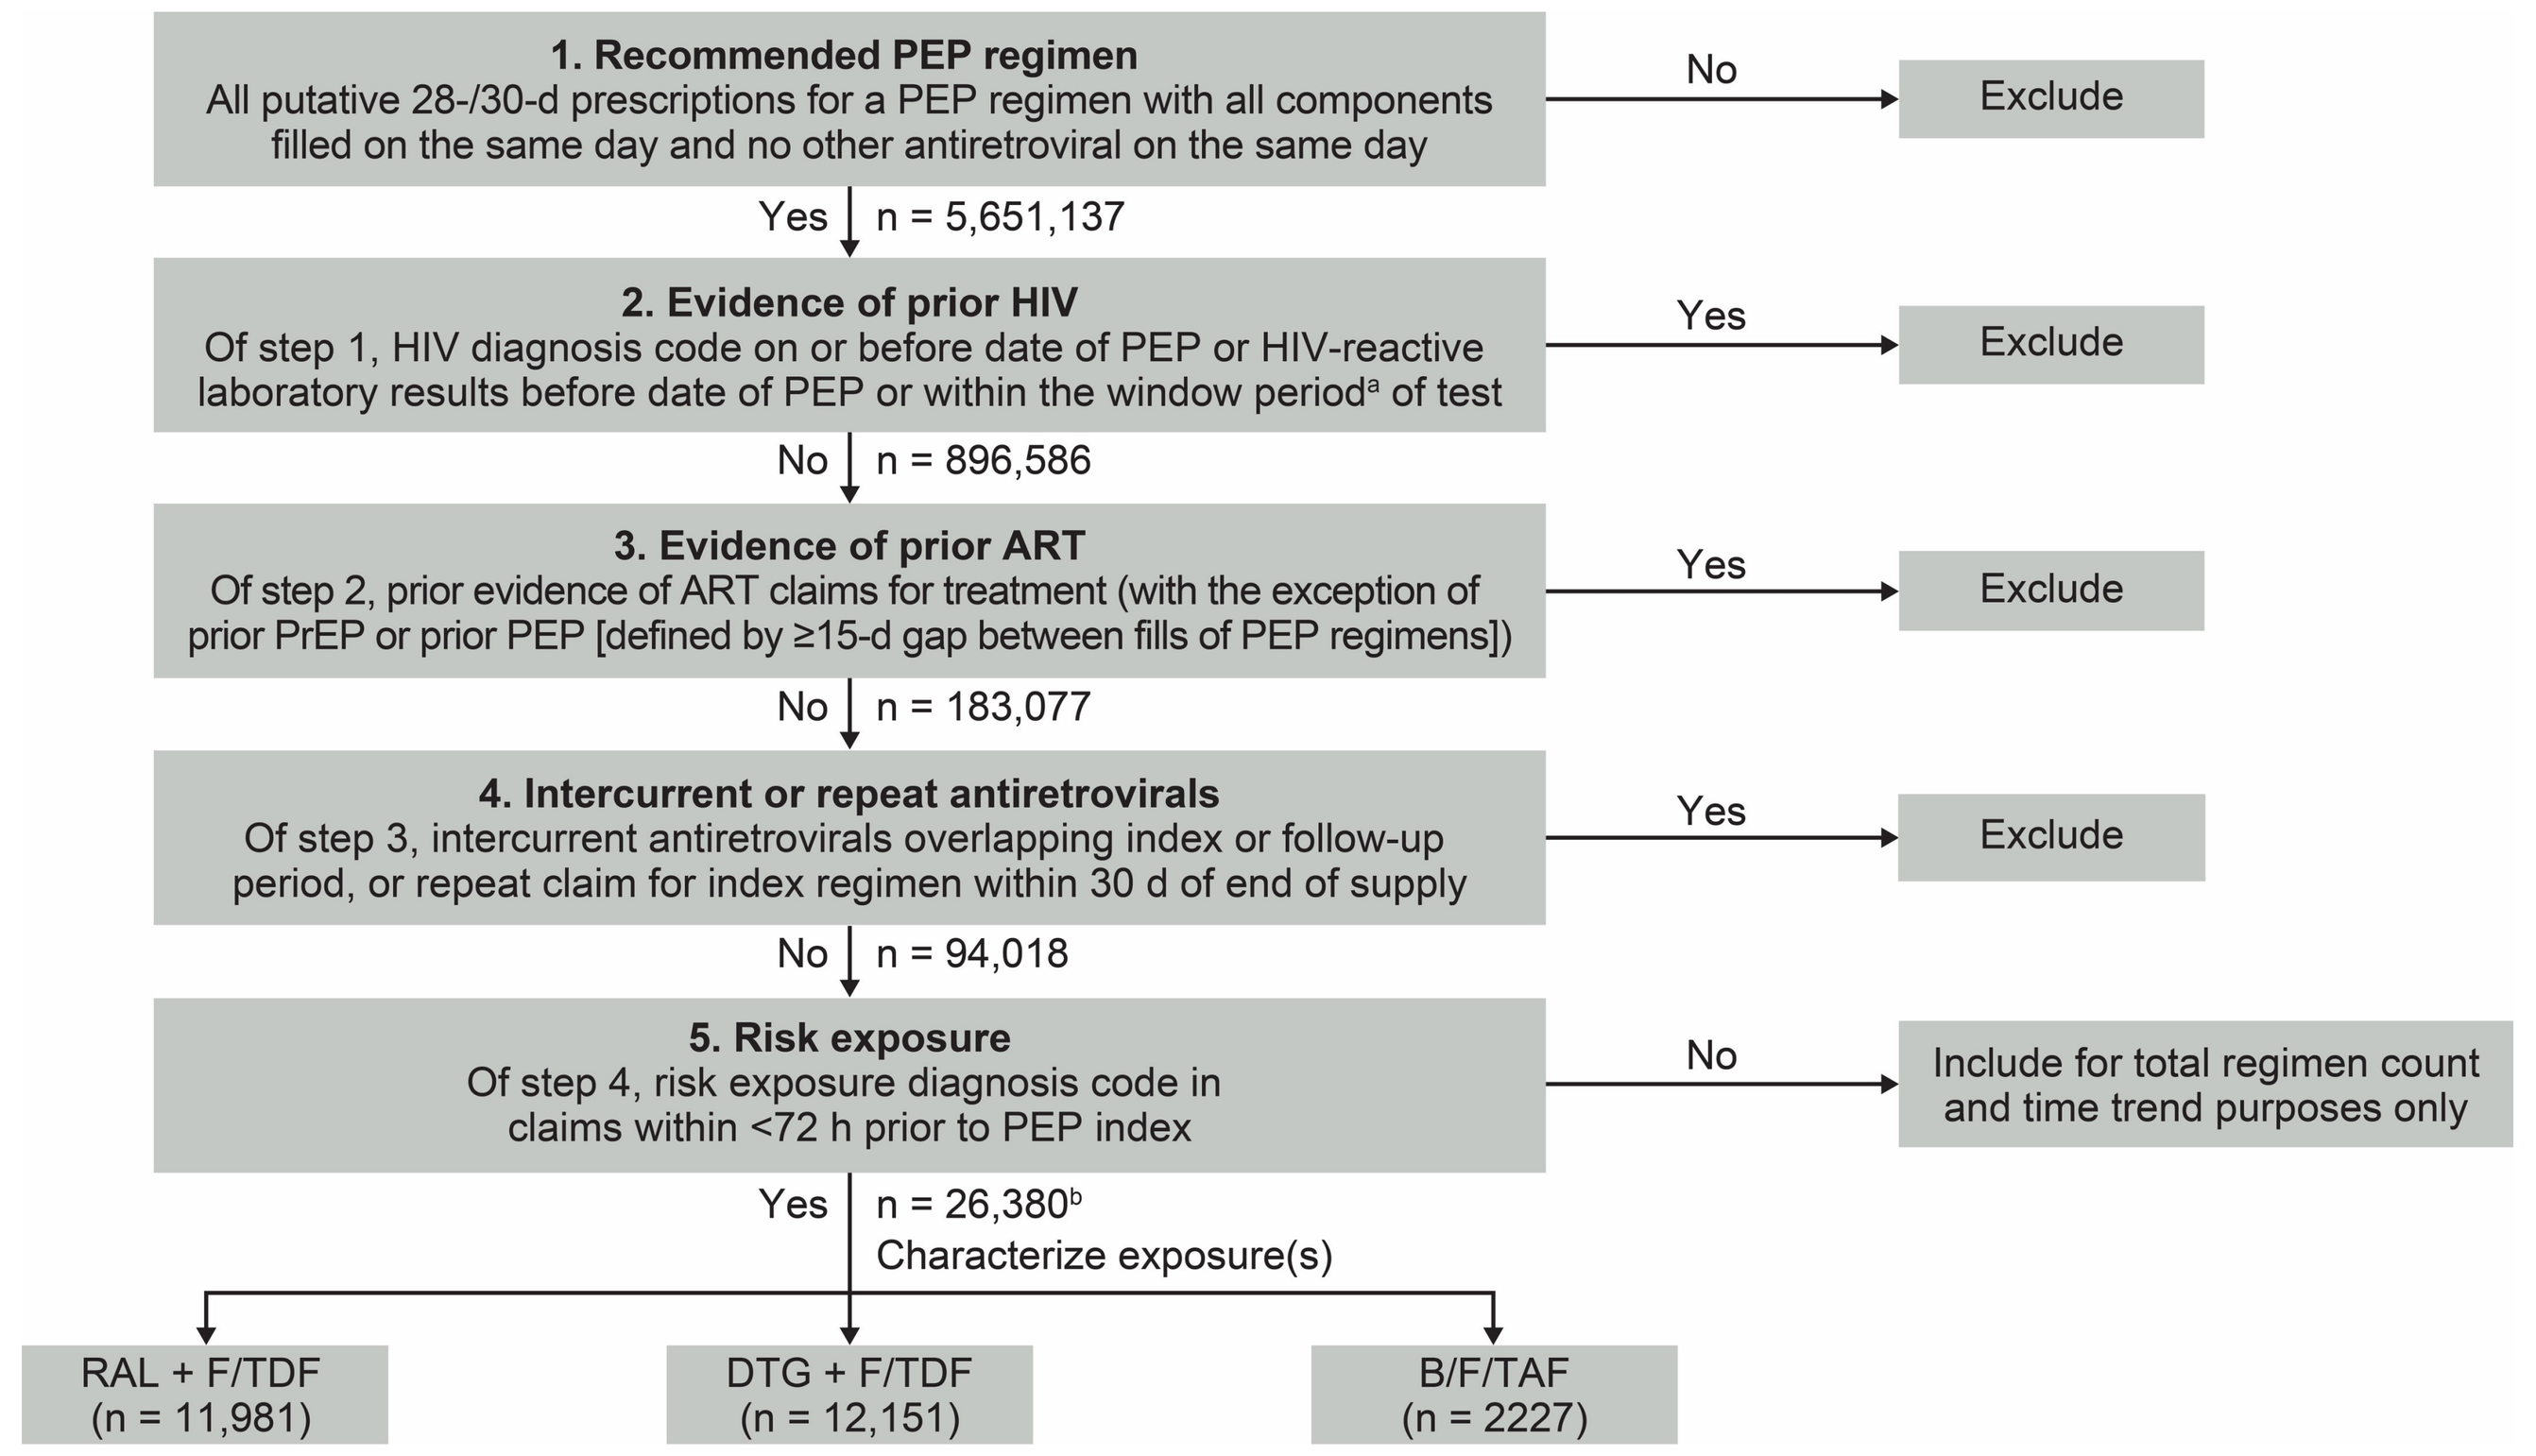

1. Recommended PEP regimen
All putative 28-/30-d prescriptions for a PEP regimen with all components filled on the same day and no other antiretroviral on the same day
No
Exclude
Yes
n = 5,651,137
2. Evidence of prior HIV
Of step 1, HIV diagnosis code on or before date of PEP or HIV-reactive laboratory results before date of PEP or within the window period[a] of test
Yes
Exclude
No
n = 896,586
3. Evidence of prior ART
Of step 2, prior evidence of ART claims for treatment (with the exception of prior PrEP or prior PEP [defined by ≥15-d gap between fills of PEP regimens])
Yes
Exclude
No
n = 183,077
4. Intercurrent or repeat antiretrovirals
Of step 3, intercurrent antiretrovirals overlapping index or follow-up period, or repeat claim for index regimen within 30 d of end of supply
Yes
Exclude
No
n = 94,018
5. Risk exposure
Of step 4, risk exposure diagnosis code in claims within <72 h prior to PEP index
No
Include for total regimen count and time trend purposes only
Yes
n = 26,380[b]
Characterize exposure(s)
RAL + F/TDF (n = 11,981)
DTG + F/TDF (n = 12,151)
B/F/TAF (n = 2227)

[a]The window periods of the laboratory tests were defined according to the CDC clinical testing guidance for HIV. The window periods were 20 days for antibody tests and 15 days for all other tests.[9]
[b]Excludes PEP prescriptions for DRV/r + F/TDF (n = 21).
ART, antiretroviral therapy; B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; CDC, Centers for Disease Control and Prevention; DRV/r + F/TDF, ritonavir-boosted darunavir + emtricitabine/tenofovir disoproxil fumarate; DTG + F/TDF, dolutegravir + emtricitabine/tenofovir disoproxil fumarate; PEP, post-exposure prophylaxis; PrEP, pre-exposure prophylaxis; RAL + F/TDF, raltegravir + emtricitabine/tenofovir disoproxil fumarate.

- Among overall PEP episodes, the majority were for individuals who were aged 25 to 44 years, were male, had unknown race/ethnicity, resided in the US South or Northeast, had commercial insurance, and had no prior PrEP use within 1 year (**Table 1**)
 - — A higher proportion of PEP prescriptions for B/F/TAF were for individuals who were aged ≥45 years, were male, and resided in the US South compared with PEP prescriptions for RAL + F/TDF and DTG + F/TDF

Table 1. Demographic Characteristics of PEP Episodes by Regimen Type[a]

Parameter, n	Total PEP (All Regimens[b]; n = 94,018)	RAL + F/TDF (n = 34,321)	DTG + F/TDF (n = 32,587)	B/F/TAF (n = 26,183)	*P* Value[c]	SMD[c]
Age group, y						
12-17	4519	2461	1452	600	<0.001	0.533
18-24	16,293	7176	6531	2550		
25-44	53,248	19,174	19,415	14,220		
45-64	18,621	5279	4989	7938		
≥65	1337	231	200	875		
Sex						
Male	54,194	15,655	19,779	18,140	<0.001	0.493
Female	39,824	18,666	12,808	8043		
Race/ethnicity						
Unknown	52,794	18,769	17,328	15,888	<0.001	0.220
Non-Hispanic White	15,359	6476	5559	3301		
Non-Hispanic Black	13,915	4705	4806	4331		
Hispanic	7171	2522	2936	1698		
Other	2903	1093	1123	681		
Asian	1876	756	835	284		
US region						
South	32,968	9470	11,523	11,666	<0.001	0.406
Northeast	27,745	12,651	8759	6000		
West	18,209	6772	7341	3982		
Midwest	15,054	5404	4958	4527		
Unknown	42	24	6	8		
Year range of index date						
2016-2017	9976	7105	2290	–[d]	<0.001	1.026
2018-2020	24,464	12,164	7899	4143		
2021-2024	50,506	13,610	18,183	18,628		
2025	9072	1442	4215	3412		
Insurance type						
Commercial	51,930	18,383	19,988	13,454	<0.001	0.235
Medicaid	31,302	12,527	10,978	7723		
Unknown	8530	2883	1190	3737		
Medicare/Medicare Advantage	2256	528	431	1269		
Any PrEP use 1 y prior to index date						
No	90,972	33,728	31,347	25,116	<0.001	0.140
Yes	3046	593	1240	1067		

[a]Data are at the PEP episode level.
[b]Total includes regimens included in CDC guidance during the study period. The 3 most common regimens are shown; the total PEP column includes PEP prescriptions for DRV/r + F/TDF (n = 927).
[c]*P* values indicate statistically significant differences in proportions by category using the chi-squared tests of independence. However, *P* values are highly significant with large sample sizes. The reported SMD represents the maximum absolute pairwise SMD among RAL + F/TDF, DTG + F/TDF, and B/F/TAF. The larger of the 2 pairwise SMD values is listed, with >0.2 indicating likely imbalance between ≥2 groups.
[d]B/F/TAF became available in February 2018.
B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; CDC, Centers for Disease Control and Prevention; DRV/r + F/TDF, ritonavir-boosted darunavir + emtricitabine/tenofovir disoproxil fumarate; DTG + F/TDF, dolutegravir + emtricitabine/tenofovir disoproxil fumarate; PEP, post-exposure prophylaxis; PrEP, pre-exposure prophylaxis; RAL + F/TDF, raltegravir + emtricitabine/tenofovir disoproxil fumarate; SMD, standardized mean difference.

PEP Risk Exposures and Prescriptions

- Among 94,018 PEP episodes, 26,359 (28%) had ≥1 HIV risk exposure code identified within <72 hours of the PEP claim
 - — The most common risk exposures overall were exposure to HIV and exposure to sexually transmitted infection (STI; **Table 2**)
- Exposure-linked B/F/TAF episodes were more frequently associated with rectal STI testing codes than exposure-linked episodes for comparator regimens
- A higher proportion of exposure-linked RAL + F/TDF episodes had exposure codes relating to contact with a contaminated hypodermic needle and sexual abuse, sexual assault, or rape compared with exposure-linked episodes for DTG + F/TDF and B/F/TAF
- Of the PEP episodes with a risk exposure code for contact with a contaminated hypodermic needle, most were presumed occupational exposures

Table 2. Risk Exposure Codes Identified Within <72 Hours of Exposure-Linked PEP Episodes[a,b]

Risk Exposure (ICD-10-CM Code), n	Total PEP (All Regimens[c]; n = 26,359)	RAL + F/TDF (n = 11,981)	DTG + F/TDF (n = 12,151)	B/F/TAF (n = 2227)	*P* Value[d]	SMD[d]
Exposure to HIV (Z20.6)	10,665	4128	5574	963	<0.001	0.235
Chlamydia and/or gonorrhea laboratory test, unspecified swab site	3908	1437	1982	489	<0.001	0.268
Chlamydia and/or gonorrhea laboratory test, rectal swab site	1853	185	1247	421	<0.001	0.598
Male	1588	123	1095	370	<0.001	0.572
Contact with contaminated hypodermic needle	3058	1903	994	161	<0.001	0.273
Substance use related	279	178	94	7	<0.001	0.124
Presumed occupational[e]	2779	1725	900	154	<0.001	0.244
Sexual abuse, sexual assault, or rape	4194	2643	1382	169	<0.001	0.416
High-risk sexual behavior, homosexual	973	275	610	88	<0.001	0.146
High-risk sexual behavior, heterosexual	2302	858	1266	178	<0.001	0.115
Exposure to viral hepatitis (Z20.5)	357	110	176	71	<0.001	0.161
Exposure to STI (Z20.2)	6969	3154	3287	528	0.00418	0.077

[a]Among those with identifiable HIV exposure information.
[b]Codes are not mutually exclusive and may have appeared in combination with other codes.
[c]Total for any PEP regimen according to CDC guidelines in 2016 or 2025. The 3 most common regimens are shown; the total PEP column excludes PEP prescriptions for DRV/r + F/TDF associated with risk exposure codes (n = 21).
[d]*P* values indicate statistically significant differences in proportions by category using chi-squared tests of independence. However, *P* values are highly significant with large sample sizes. The reported SMD represents the maximum absolute pairwise SMD among RAL + F/TDF, DTG + F/TDF, and B/F/TAF. The larger of the 2 pairwise SMD values is listed, with >0.2 indicating likely imbalance between ≥2 groups.
[e]Contact with contaminated hypodermic needle exposure codes were presumed occupational based on the absence of a substance use–related exposure code.
B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; CDC, Centers for Disease Control and Prevention; DRV/r + F/TDF, ritonavir-boosted darunavir + emtricitabine/tenofovir disoproxil fumarate; DTG + F/TDF, dolutegravir + emtricitabine/tenofovir disoproxil fumarate; ICD-10-CM, *International Classification of Diseases, Tenth Revision, Clinical Modification*; PEP, post-exposure prophylaxis; RAL + F/TDF, raltegravir + emtricitabine/tenofovir disoproxil fumarate; SMD, standardized mean difference; STI, sexually transmitted infection.

- PEP prescriptions for RAL + F/TDF peaked in 2019 before declining and plateauing (**Figure 2**)
- PEP prescriptions for DTG + F/TDF have risen steadily since 2016, with the exception of a drop in 2020
- The use of B/F/TAF as PEP has steadily increased since 2018, even prior to updated CDC guidelines recommending it for PEP; interpretation should consider that B/F/TAF entered the market later than comparator regimens

Figure 2. PEP Prescriptions Over Time by Regimen Type[a]

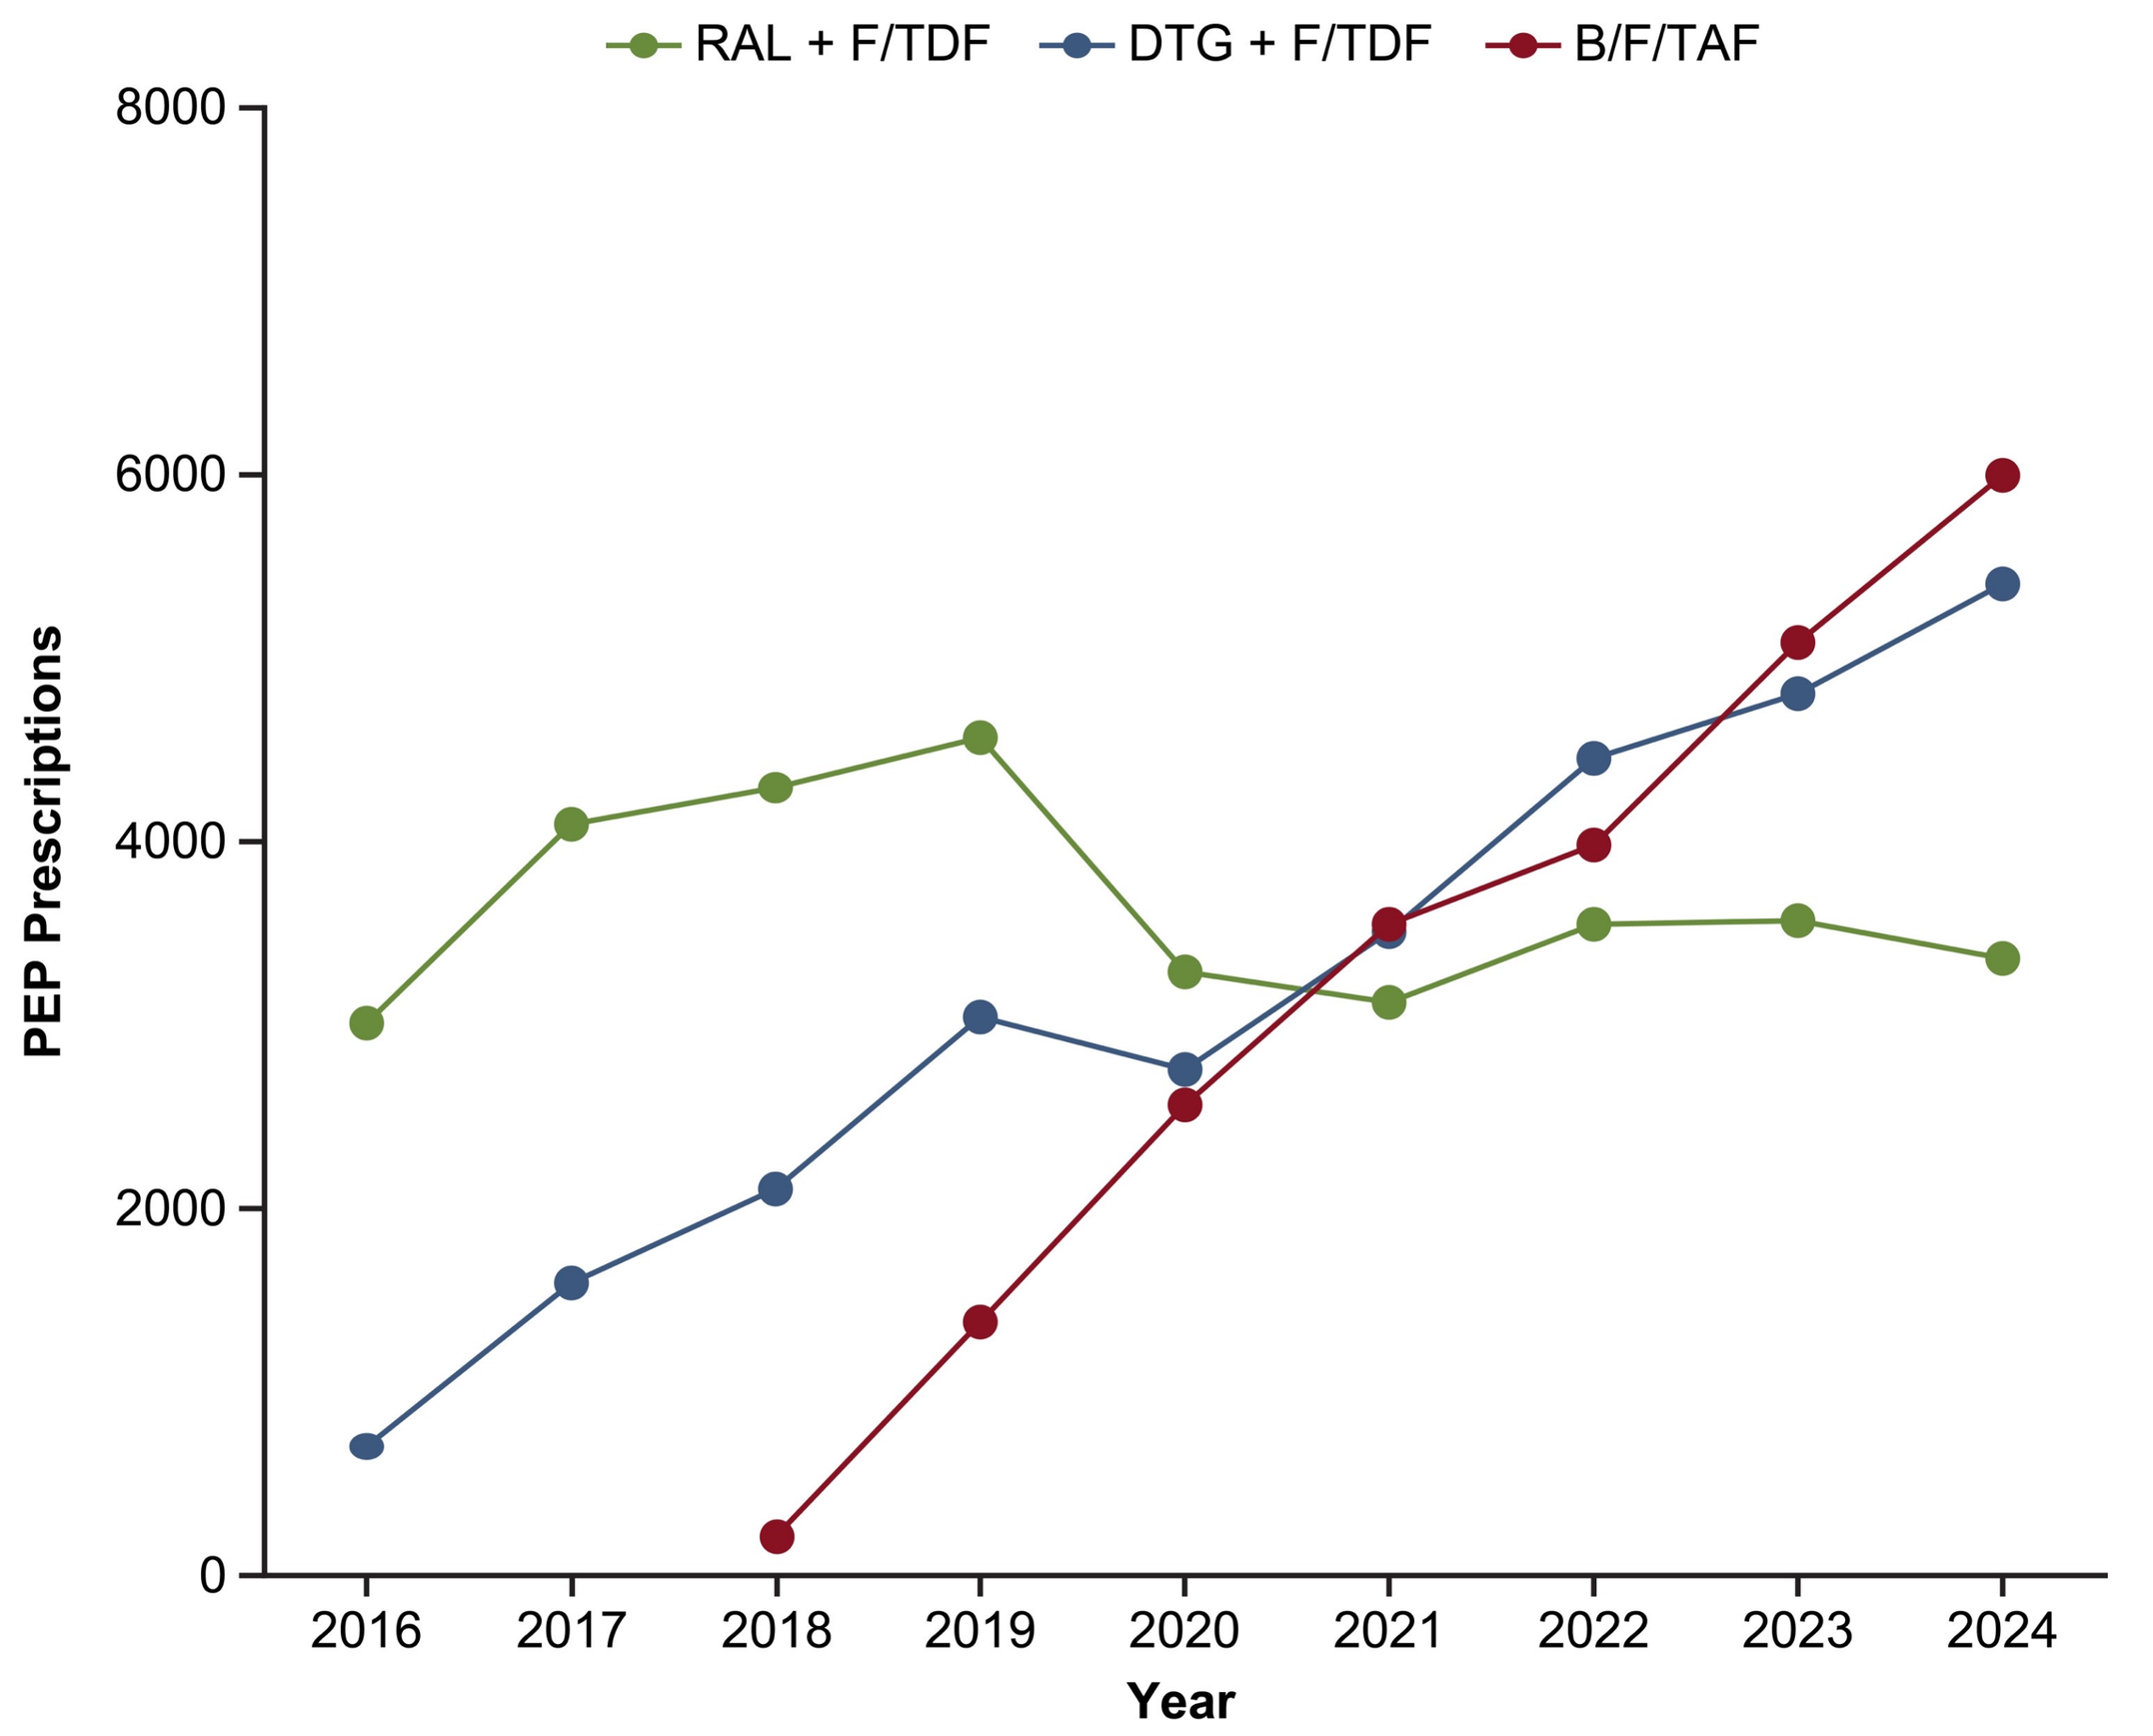

RAL + F/TDF
DTG + F/TDF
B/F/TAF
8000
6000
4000
2000
0
PEP Prescriptions
2016
2017
2018
2019
2020
2021
2022
2023
2024
Year

[a]PEP prescription data from 2020 to 2021 may have been impacted by COVID-19. A partial year of 2025 data was excluded from the time trend analysis to avoid misinterpretation as a decline in 2025.
B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; DTG + F/TDF, dolutegravir + emtricitabine/tenofovir disoproxil fumarate; PEP, post-exposure prophylaxis; RAL + F/TDF, raltegravir + emtricitabine/tenofovir disoproxil fumarate.

Post-PEP HIV Diagnoses

- HIV diagnoses or reactive HIV test results during Days 28 to 42 following PEP were rare among individuals with documented HIV-negative baseline testing (**Table 3**)

Table 3. HIV Diagnoses Following PEP by Regimen

Regimen	HIV Diagnoses Following PEP, n/N (%)
RAL + F/TDF (n = 4447)	4/4447 (0.09)
DTG + F/TDF (n = 4999)	5/4999 (0.10)
B/F/TAF (n = 949)	1/949 (0.11)

New HIV infections after PEP with documented evidence that the individual was negative for HIV at baseline (–3 to +10 days relative to the index date) were included.
B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; DTG + F/TDF, dolutegravir + emtricitabine/tenofovir disoproxil fumarate; PEP, post-exposure prophylaxis; RAL + F/TDF, raltegravir + emtricitabine/tenofovir disoproxil fumarate.

Limitations

- Risk exposure data were not available for all evaluated PEP claims
- Pharmacy claims confirm medication dispensing but cannot confirm treatment initiation, adherence, or completion
- Definitive HIV diagnoses following PEP use may have been related to subsequent risk exposures rather than the original risk exposure for which PEP was sought
- As this analysis was based on insurance claims data, PEP use by uninsured individuals was not captured, which may have excluded potentially high-risk populations
- Comparisons of exposure type distributions across regimens may be influenced by calendar-time confounding and differential coding practices over the study period
- Observed differences between regimen groups should be interpreted in the context of changing PEP prescribing practices over time and are not intended to represent comparative effectiveness or causal associations
- HIV exposure codes and laboratory testing codes were used as proxy measures of exposure circumstances and may not fully represent the actual indication for PEP prescribing
- The diagnosis codes Z20.6, Z20.5, and Z20.2 indicate exposure to HIV, viral hepatitis, and STI, respectively, which may be self-reported by the patient without external verification, similar to real-world PEP encounters
- While the HIV diagnosis codes employed indicate documentation of HIV status by the health care provider, they do not necessarily indicate the original date of infection or require a confirmatory laboratory result at or around the time of the claim

References: **1.** Fauci AS, et al. *JAMA*. 2019;321:844-5. **2.** Tanner MR, et al. *MMWR Recomm Rep*. 2025;74:1-56. **3.** Mayer KH, Allan-Blitz LT. *Lancet HIV*. 2023;10:e816-24. **4.** Allan-Blitz LT, Mayer KH. *Open Forum Infect Dis*. 2024;11:ofae332. **5.** Ford N, et al. *AIDS*. 2014;28:2721-7. **6.** John SA, et al. *AIDS Behav*. 2020;24:3124-31. **7.** Jain S, et al. *Clin Infect Dis*. 2015;60:S200-4. **8.** Le JN, et al. *Clin Infect Dis*. 2026;82:e286-95. **9.** Centers for Disease Control and Prevention. Clinical testing guidance for HIV. Accessed September 15, 2026. https://www.cdc.gov/hivnexus/hcp/diagnosis-testing/index.html.

Acknowledgments: This study was funded by Gilead Sciences, Inc. Medical writing and editorial support were provided by Jane Abolafia, PhD, of Lumanity Communications Inc., and were funded by Gilead Sciences, Inc.

Disclosures: **KHM** received grant/research support from Gilead Sciences, Inc., Merck, and ViiV Healthcare. **TL**, **VS**, **WW**, and **JTH** are stockholders and employees of Gilead Sciences, Inc. **LMM** works exclusively on Gilead Sciences, Inc., studies.

IDWeek; October 21–24, 2026; Washington, DC, USA